Infigratinib versus gemcitabine plus cisplatin multicenter, open-label, randomized, phase 3 study in patients with advanced cholangiocarcinoma with *FGFR2* gene fusions/translocations: the PROOF trial

Javle M,[1] Borbath I,[2] Clarke S,[3] Hitre E,[4] Louvet C,[5] Macarulla T,[6] Oh D-Y,[7] Spratlin J,[8] Valle JW,[9] Weiss KH,[10] Berman C,[11] Howland M,[11] Ye Y,[11] Cho T,[11] Moran S,[11] Abou-Alfa GK[12]

[1]MD Anderson Cancer Center, Houston, TX; [2]Cliniques Universitaires St Luc Bruxelles, Bruxelles, Belgium; [3]University of Sydney, Sydney, Australia; [4]National Institute of Oncology, Budapest, Hungary; [5]Institut Mutualiste Montsouris, Paris, France; [6]Hospital Vall D'Hebron, Barcelona, Spain; [7]Seoul National University Hospital, Seoul, Korea, Republic of (South); [8]Alberta Health Services, Edmonton, AB; [9]University of Manchester / The Christie, Manchester, United Kingdom; [10]University Hospital Heidelberg, Heidelberg, Germany; [11]QED Therapeutics Inc, San Francisco, CA; [12]Memorial Sloan Kettering Cancer Center, New York, NY

#TPS4155

Background

- Cholangiocarcinoma is the most common biliary tract malignancy with an estimated incidence in Europe of 0.4–1.8/100,000 patients, and approximately 5,000–10,000 new cases annually in the USA.
- The fibroblast growth factor receptor (FGFR) family plays an important role in cholangiocarcinoma.
- *FGFR* translocations (i.e. fusion events) represent driver mutations in cholangiocarcinoma. They are present in 13–17% of intrahepatic cholangiocarcinomas and may predict tumor sensitivity to FGFR inhibitors.[1–3]
- Infigratinib (BGJ398), an ATP-competitive FGFR1–3-selective oral tyrosine kinase inhibitor (Figure 1), has shown clinical activity against tumors with *FGFR* alterations.[4]
- Based on earlier response data of infigratinib in relapsed/refractory cholangiocarcinoma with *FGFR2* fusions/translocations (phase 2 study CBJG398X2204),[5,6] the PROOF trial is evaluating infigratinib versus gemcitabine + cisplatin in front-line patients with advanced cholangiocarcinoma with *FGFR2* gene fusions/translocations.

Figure 1. Infigratinib: an oral FGFR1–3 selective tyrosine kinase inhibitor

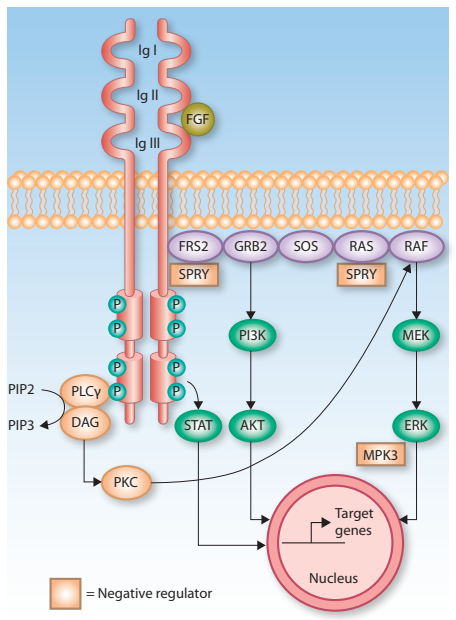
Ig I
Ig II
Ig III
FGF
FRS2
GRB2
SOS
RAS
RAF
SPRY
SPRY
P
PI3K
MEK
PIP2
PIP3
PLCγ
DAG
STAT
AKT
ERK
MPK3
PKC
Target genes
Nucleus
= Negative regulator

- Infigratinib is an orally-available, selective, ATP-competitive FGFR1–3 inhibitor
- Infigratinib has proven activity in tumor models with *FGFR* alterations

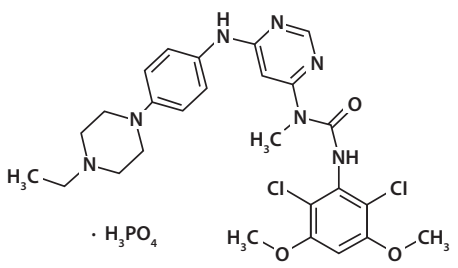
· H_3PO_4

Biochemical activity (nM IC_{50})	
FGFR1	**1.1**
FGFR2	**1.0**
FGFR3	**2.0**
FGFR4	**61.0**
VEGFR2	**210.0**

PROOF study design

- PROOF is a multicenter, open-label, randomized, controlled phase 3 study to evaluate the efficacy of infigratinib vs gemcitabine + cisplatin in patients with advanced/metastatic or inoperable cholangiocarcinoma with *FGFR2* fusions/translocations.
- Clinicaltrials.gov identifier: NCT03773302
- PROOF is a multicenter study involving approximately 145 sites worldwide.

Figure 2. PROOF study design

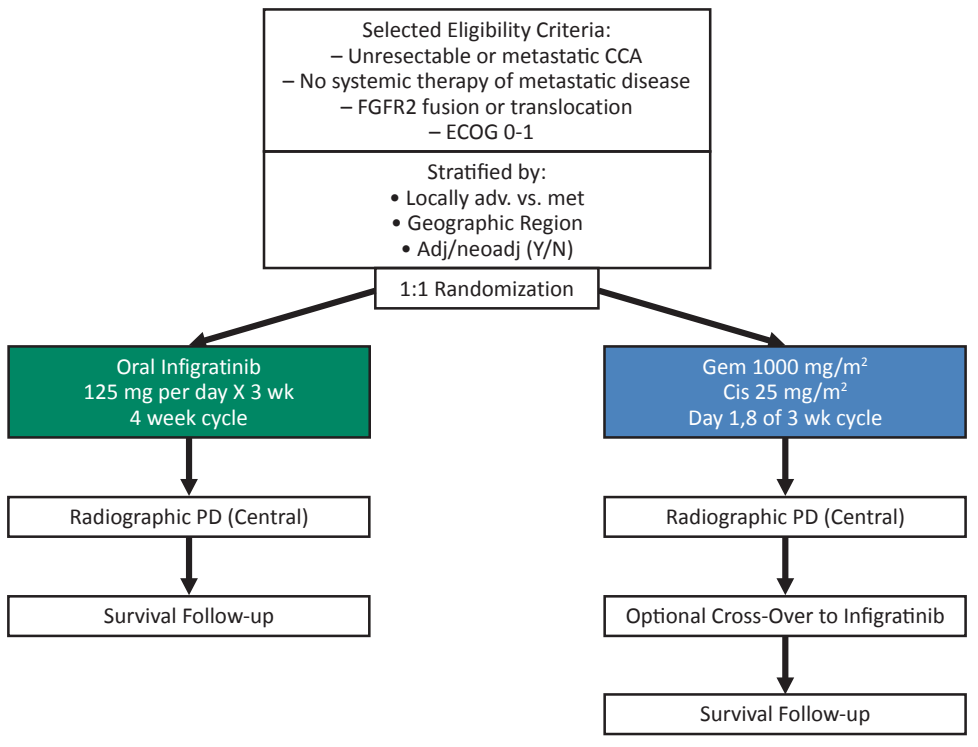
Selected Eligibility Criteria:
– Unresectable or metastatic CCA
– No systemic therapy of metastatic disease
– FGFR2 fusion or translocation
– ECOG 0-1
Stratified by:
• Locally adv. vs. met
• Geographic Region
• Adj/neoadj (Y/N)
1:1 Randomization
Oral Infigratinib
125 mg per day X 3 wk
4 week cycle
Gem 1000 mg/m²
Cis 25 mg/m²
Day 1,8 of 3 wk cycle
Radiographic PD (Central)
Radiographic PD (Central)
Survival Follow-up
Optional Cross-Over to Infigratinib
Survival Follow-up

Study objectives

- **Primary objective:** determine if infigratinib improves centrally assessed PFS vs gemcitabine + cisplatin in patients with advanced/metastatic or inoperable cholangiocarcinoma with *FGFR2* gene fusions/translocations.
- **Secondary objectives:**
 - Evaluate efficacy of infigratinib vs gemcitabine + cisplatin in terms of overall survival (OS).
 - Evaluate efficacy of infigratinib vs gemcitabine + cisplatin in terms of investigator-assessed PFS and overall response rate (ORR) based on central and investigator assessments.
 - Further evaluate efficacy in patients treated with these regimens in terms of best overall response (BOR), duration of response and disease control rate determined centrally and by the investigator.
 - Characterize the safety and tolerability of single-agent infigratinib.
- **Exploratory objectives:**
 - Compare quality of life (QOL) in patients treated with these regimens.
 - Calculate selected pharmacokinetic parameters for infigratinib.
 - Evaluate correlation between co-existing mutations, including molecular testing, and response rate of clinical endpoints.

Key inclusion criteria

- Histologically or cytologically confirmed non-resectable, recurrent, or metastatic cholangiocarcinoma. Patients with gallbladder cancer or ampullary carcinoma are not eligible.
- Written documentation of local or central laboratory determination of FGFR2 gene fusions/translocations.
- A representative tumor sample available for central FGFR2 fusion/translocation molecular testing.
- Full recovery from prior surgery, adjuvant radiotherapy or chemotherapy, and photodynamic treatment.
- Age ≥18 years.
- Eastern Cooperative Oncology Group (ECOG) performance status ≤ 1.
- Life expectancy >3 months.
- Recovery from adverse events (AEs) associated with previous systemic anti-cancer therapies to baseline or Grade 1, except for alopecia.

Key exclusion criteria

- Treatment with any systemic anti-cancer therapy for unresectable, recurrent, or metastatic cholangiocarcinoma. Prior neoadjuvant or adjuvant therapy is permitted if completed >6 months prior to first dose of study drug.
- History of a liver transplant.
- Prior or current treatment with a MEK or selective FGFR inhibitor.
- History of another primary malignancy within 3 years except adequately treated in situ carcinoma of the cervix or non-melanoma carcinoma of the skin or any other curatively treated malignancy that is not expected to require treatment for recurrence during the course of the study.
- Impairment of gastrointestinal (GI) function or GI disease that may significantly alter the absorption of oral infigratinib (e.g., ulcerative diseases, uncontrolled nausea, vomiting, diarrhea, malabsorption syndrome, small bowel resection).

Treatment

- Patients are randomized 1:1 to oral infigratinib once daily for 21 days of a 28-day treatment cycle versus IV gemcitabine (1000 mg/m2) + cisplatin (25 mg/m2) on days 1 and 8 of a 21-day cycle.
- Treatment will continue until confirmed progressive disease by central review, intolerance, withdrawal of informed consent, or death.
- After 8 cycles of gemcitabine + cisplatin, patients can continue treatment if the investigator considers that they are deriving continued benefit.
- Patients on the gemcitabine + cisplatin arm with central radiographic progression can cross-over to infigratinib.

Dose modifications/treatment delays

- Patients who do not tolerate the protocol-specified dosing schedule are permitted to have dose adjustments in order to allow continuation of study drug.
- Each patient is allowed up to two dose reductions (Table 1).
- Patients should discontinue infigratinib if toxicities persist following two dose reductions, unless discussed and approved by the QED Therapeutics' Medical Monitor.

Table 1. Dose-reduction scheme for infigratinib

Dose reduction	Starting dose level 0	Dose level –1	Dose level –2
Infigratinib	125 mg	100 mg	75 mg

Efficacy evaluation

- Tumor response will be evaluated by independent central review and by the investigator according RECIST Version 1.1.[7]
- Patient management will be based upon investigator evaluations; patients should remain on study drug until central confirmation of progressive disease.
- Survival status and use of new anticancer medications will be followed approximately every 3 months once progressive disease has been documented and centrally confirmed.
- Survival status and use of anticancer therapy will be followed up to 1 year after the time at which 251 centrally confirmed PFS events are reached (i.e. end of study).

Safety evaluation

- Safety evaluation will be based on AE reporting, laboratory parameters, vital signs, physical examinations, 12-lead ECGs, cardiac imaging, and ophthalmic assessments.
- Tolerability will be assessed by the incidence of AEs leading to study drug interruption, dose reduction, or discontinuation.

Quality of life evaluation

- QOL will be evaluated using the EQ-5D, which measures 5 dimensions of health, including mobility, self-care, usual activities, pain/discomfort, anxiety, and general health; the EORTC QLQ-C30, a reliable and valid measure of QOL in cancer patients; and the EORTC QLQ-BIL21, a disease-specific module for patients with cholangiocarcinoma and gallbladder cancer.

Pharmacokinetics (infigratinib group only)

- On the days of PK sampling, patients should bring their study drug with them to the clinic and take it immediately after the pre-dose PK sample is taken.
- The PK parameters C_{ss}, AUC_{ss}, CL/F, and R_{acc} will be calculated for infigratinib, and the concentrations of the metabolites will be evaluated. For patients in the infigratinib group who discontinue study drug, attempts should be made to collect a PK blood sample from the patient at the time of discontinuation.

Biomarkers

- Archival or newly obtained tumor samples will be collected to explore mechanisms of resistance to cancer treatment using next generation sequencing analysis.
- Blood samples will also be collected for assessment of cell-free DNA.

Planned patient population and current status

Planned sample size/statistics

- Approximately 350 patients who have tumors with confirmed *FGFR2* gene fusions/translocations by a central laboratory (175 patients per group) are planned for study participation.
- Assuming a PFS hazard ratio (HR) of 0.7 comparing infigratinib to gemcitabine with cisplatin, the study will provide approximately 80% power to demonstrate that infigratinib improves the centrally assessed PFS compared to treatment with gemcitabine and cisplatin at a 2-sided significance level of 0.05.
- The study employs a group sequential design with one interim analysis on PFS, which will be conducted when approximately 33% of the PFS events are observed. The primary analysis for PFS will be conducted after approximately 251 PFS events have been observed.

Current status

- The study was initiated in February 2019 and is projected to reach the planned number of PFS events in approximately 32 months from randomization of the first subject.

Acknowledgements

Editorial/writing support for this poster was provided by Miller Medical Communications Ltd. This work was funded by the study sponsor (QED Therapeutics).

References

1. Graham RP, et al. Hum Pathol 2014;45:1630–8.
2. Ross JS, et al. Oncologist 2014;19:235–42.
3. Farshidfar F, et al. Cell Rep 2017;18:2780–94.
4. Guagnano V, et al. Cancer Discov 2012;2:1118–33.
5. Javle M, et al. Proc APCCA 2019 (poster P-19).
6. Javle M, et al. J Clin Oncol 2018;36:276–82.
7. Eisenhauer EA, et al. Eur J Cancer 2009;45:228–47.

Presented at the 2019 ASCO Annual Meeting, May 31 – June 4, Chicago, Illinois, USA

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors